Real-world treatment patterns and clinical outcomes in patients with metastatic urothelial carcinoma (mUC): results of a nationwide, longitudinal, retrospective study in Hungary

A. Maráz,[1] B. Nagy,[2] T. Macher,[2] J. Jeskó,[2] E. Tischler,[3] Cs. Csongvai,[3] M. Kearney[4]

[1]Department of Oncotherapy, University of Szeged, Szeged, Hungary; [2]Healthware Consulting Ltd., Budapest, Hungary; [3]Merck Kft., Budapest, Hungary, an affiliate of Merck KGaA; [4]Merck Healthcare KGaA, Darmstadt, Germany

SCOPE

- **This study aimed to describe basic epidemiology, patient characteristics, and overall survival (OS) in patients with mUC treated with first-line (1L) chemotherapy or immuno-oncology (IO) monotherapy in a real-world setting in Hungary, and thus provide relevant and contemporary information to healthcare providers, patients and their families, payers, and other stakeholders**

CONCLUSIONS

- **This first-of-its-kind, nationwide, descriptive, retrospective real-world study provides insights into treatment patterns for mUC in routine clinical practice in Hungary**
- **Only half of all patients with mUC in this study received systemic treatment. Of those who did, the majority (86.1%) received guideline-recommended 1L platinum-based chemotherapy (PBC). IO use was limited (6.1%) because of its more recent approval and restricted reimbursement**
- **Clinical outcomes associated with 1L agents and nontreatment were consistent with previously published real-world data[1-4]**
- **Among those with 1L treatment, real-world median OS was longer in patients receiving 1L PBC than in patients receiving other 1L treatment options studied (ie, 12.8, 7.5, and 6.3 months with 1L PBC, 1L non-PBC, and 1L IO, respectively)**
- **Future research should evaluate unmet needs, reasons for undertreatment, and the impact of limited access to IO, and outcomes should be assessed when the recently approved 1L IO maintenance treatment is reimbursed in Hungary**

GET POSTER PDF

Copies of this poster obtained through this quick response (QR) code are for personal use only and may not be reproduced without permission from ESMO and the author of this poster.

Correspondence: **Anikó Maráz**, dr.aniko.maraz@gmail.com

BACKGROUND

- UC is the most common malignancy involving the urinary system and is the fourth most common tumor in developed countries[5]
 - UC is over 4 times more common in men than women, with an incidence of 9.6 in 100,000 in men and 2.4 in 100,000 in women worldwide[6,7]
- In Hungary in 2018, the age-standardized incidence and mortality rates for all ages and sexes were estimated at 16.9 in 100,000 and 3.6 in 100,000, respectively[7,8]; however, no standardized local data for patients with mUC have been published, and data describing the treatment patterns for mUC in routine clinical practice in Eastern Europe are scarce
- Treatment regimens for mUC remained relatively unchanged until the emergence of immune checkpoint inhibitors[9]
- As outlined in current international guidelines,[10,11] PBC is a standard 1L treatment for patients with mUC, with patients receiving cisplatin or carboplatin + gemcitabine depending on eligibility[12]
- 1L IO was approved by the European Commission in 2017 and accepted into the Named-Patient Based Reimbursement (NPBR) program in Hungary in 2018

METHODS

- This study consisted of a retrospective analysis of de-identified patient data from the National Health Insurance Fund Administration (NHIFA) database, the only health insurance fund in Hungary covering all Hungarian inhabitants (approximately 10 million)[13]
- The study period was from 1 January 2016 through 30 June 2021, with a 1-year censor period from 1 January 2015
- The cohort of patients with mUC were identified as those with *International Classification of Diseases*, *Tenth Revision* (*ICD-10*) codes C65-C68, denoting malignant neoplasms of urinary tract, who had evidence of metastatic disease (either ICD-10 codes C77-C79 or relevant mUC 1L treatment) (**Figures 1 and 2**)
- Patients were categorized into 2 cohorts: untreated and treated. The treated cohort was subdivided by the type of 1L treatment received: (1) PBC, (2) non-PBC, or (3) IO monotherapy per label (**Figure 1**)
- Descriptive statistical analyses were conducted to summarize the results
 - The Kaplan-Meier method was used to estimate median OS (R 4.0.4 survival package)
 - Unadjusted median OS was calculated from Index date (1L treatment cohort: start date of 1L systemic treatment; untreated cohort: date of mUC diagnosis)
- Ethics approval, as required by Ministerial decree no. 23/2002 (V.9) for non interventional studies, was provided by the Hungarian Medical Research Council (No: IV/7775-4/2021/EKU)[14]

Figure 1. Patient Attrition

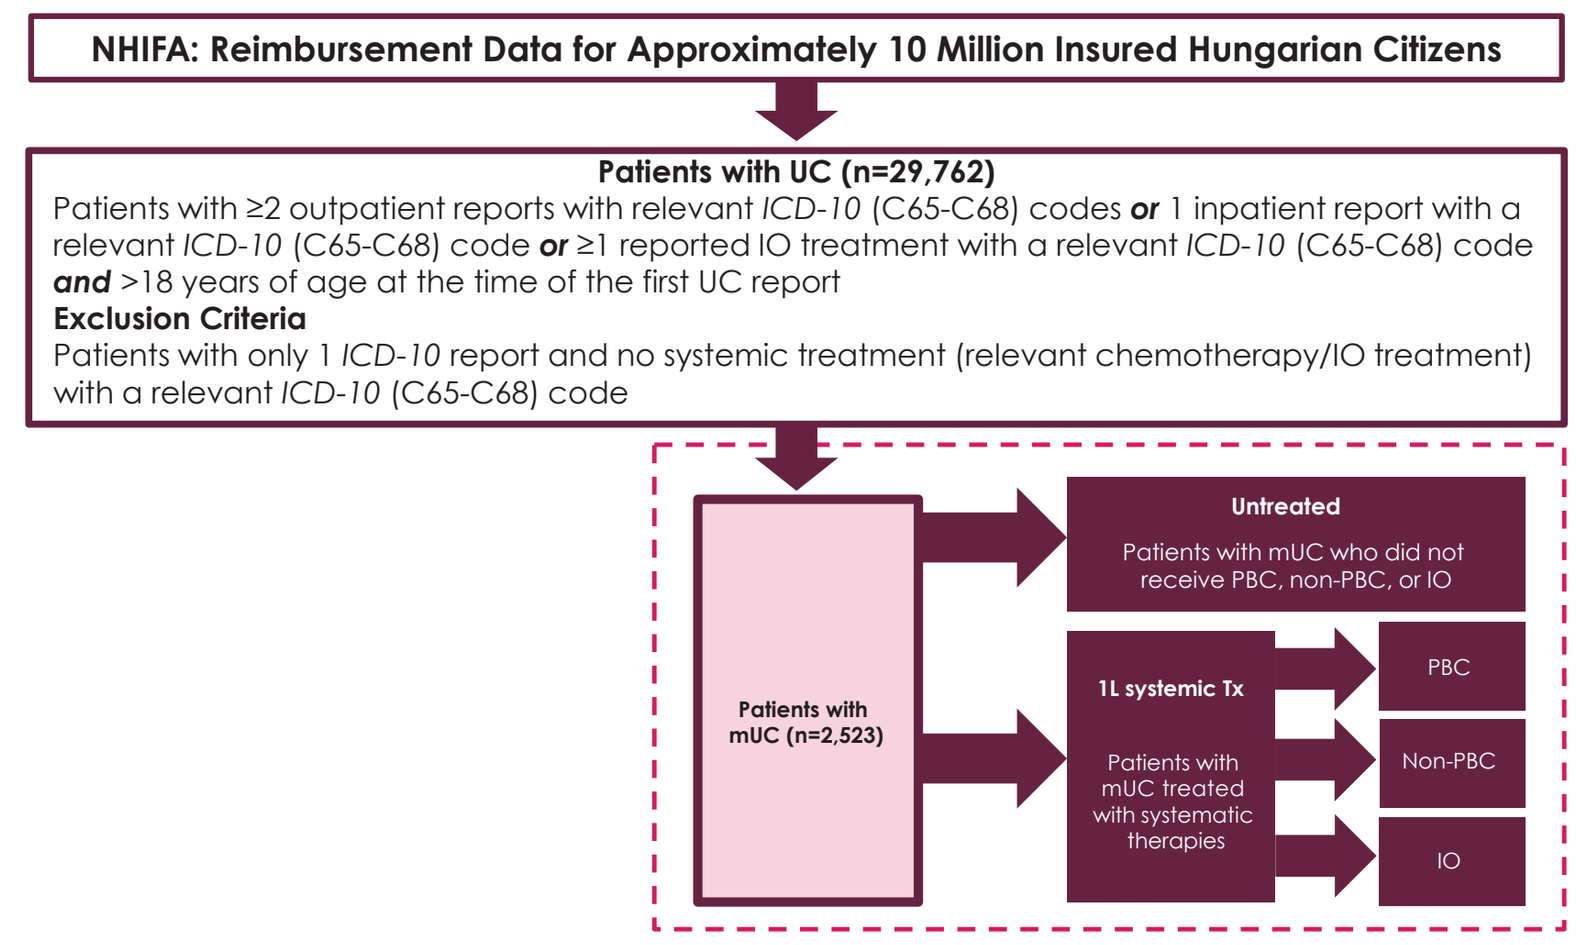
NHIFA: Reimbursement Data for Approximately 10 Million Insured Hungarian Citizens
Patients with UC (n=29,762)
Patients with ≥2 outpatient reports with relevant ICD-10 (C65-C68) codes or 1 inpatient report with a relevant ICD-10 (C65-C68) code or ≥1 reported IO treatment with a relevant ICD-10 (C65-C68) code and >18 years of age at the time of the first UC report
Exclusion Criteria
Patients with only 1 ICD-10 report and no systemic treatment (relevant chemotherapy/IO treatment) with a relevant ICD-10 (C65-C68) code
Patients with mUC (n=2,523)
Untreated
Patients with mUC who did not receive PBC, non-PBC, or IO
1L systemic Tx
Patients with mUC treated with systemic therapies
PBC
Non-PBC
IO

IO, immuno-oncology; **NHIFA**, National Health Insurance Fund Administration; **mUC**, metastatic urothelial carcinoma; **non-PBC**, non–platinum-based chemotherapy; **PBC**, platinum-based chemotherapy; **ICD-10**, *International Classification of Diseases, Tenth Revision*; **Tx**, treatment; **UC**, urothelial carcinoma.

Figure 2. Study Design

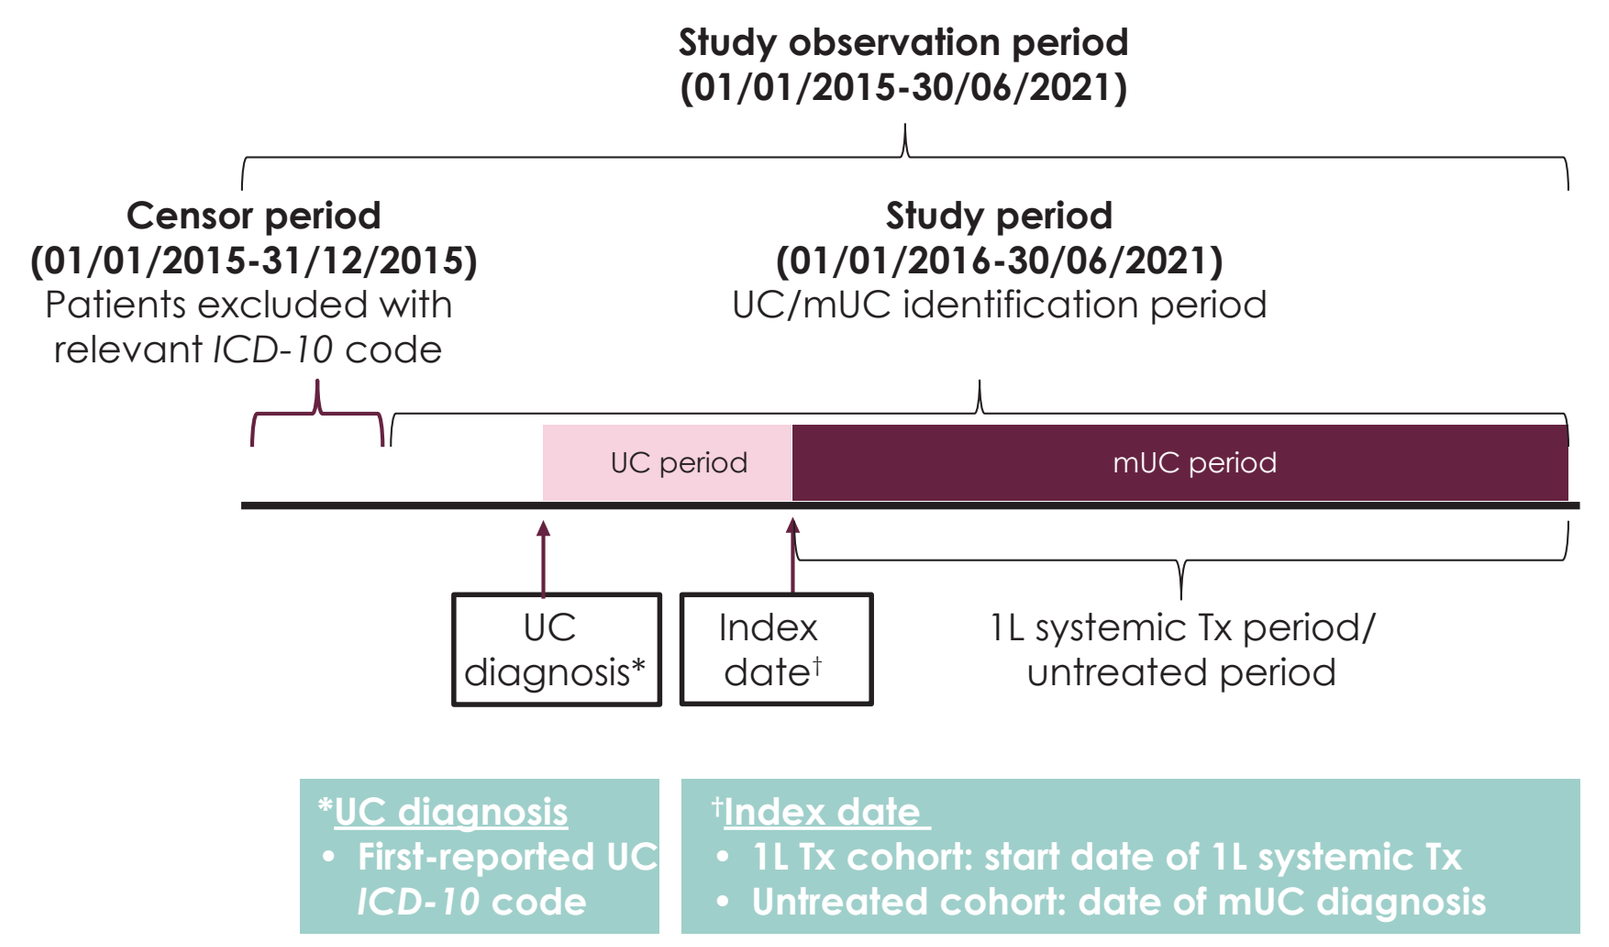
Study observation period
(01/01/2015-30/06/2021)
Censor period
(01/01/2015-31/12/2015)
Patients excluded with relevant ICD-10 code
Study period
(01/01/2016-30/06/2021)
UC/mUC identification period
UC period
mUC period
UC diagnosis*
Index date†
1L systemic Tx period/ untreated period
*UC diagnosis
• First-reported UC ICD-10 code
†Index date
• 1L Tx cohort: start date of 1L systemic Tx
• Untreated cohort: date of mUC diagnosis

1L, first line; **ICD-10**, *International Classification of Diseases, Tenth Revision*; **IO**, immuno-oncology; **mUC**, metastatic urothelial carcinoma; **Tx**, treatment; **UC**, urothelial carcinoma.

RESULTS

Patient Characteristics

- A total of 2,523 patients with mUC met the selection criteria; the majority (72.2%) were male with a median age of 67 years, which was consistent between treated and untreated patients
 - In the IO-treated cohort, the mean and median ages of patients were higher than those of the overall population (**Table 1**)

Treatment Patterns

- 1,256 patients (49.8%) had an identified 1L systemic treatment, and 1,267 (50.2%) had no identified 1L treatment (**Figure 3**)
 - Among treated patients, 1L treatment was PBC in 86.1% (n=1,082), non-PBC in 7.7% (n=97), and IO in 6.1% (n=77); IO use was limited as it only became available by the NPBR program later in the study period (since 2018)

Comorbidities

- In the overall population, the most frequently occurring comorbidities during the study period were diseases of the circulatory system (93.3% of patients had ≥1 relevant reported event at any site of care in the NHIFA database)
- The comorbidity rate was higher in the IO-treated population compared with the total patient population (**Table 2**)

Table 1. Demographic characteristics at index date*

Baseline characteristics	Total patients (n=2,523)	Untreated (n=1,267)	1L systemic treatment (n=1,256)	1L treatment cohorts		
				PBC (n=1,082)	Non-PBC (n=97)	IO (n=77)
Male, n (%)	1,822 (72.2)	912 (72.0)	910 (72.5)	797 (73.7)	65 (67.0)	48 (62.3)
Mean age, years	67.3	67.8	66.8	66.6	67.1	70.2
Median age, years	67	68	67	67	66	70
Standard deviation	8	9	8	8	8	7
IQR	63-73	62-74	63-73	63-72	63-72	65-75

1L, first line; **IO**, immuno-oncology; **IQR**, interquartile range; **non-PBC**, non–platinum-based chemotherapy; **PBC**, platinum-based chemotherapy.
*Index date: 1L systemic treatment cohort at start date of 1L systemic treatment; untreated cohort at date of mUC diagnosis.

Figure 3. Treatment patterns

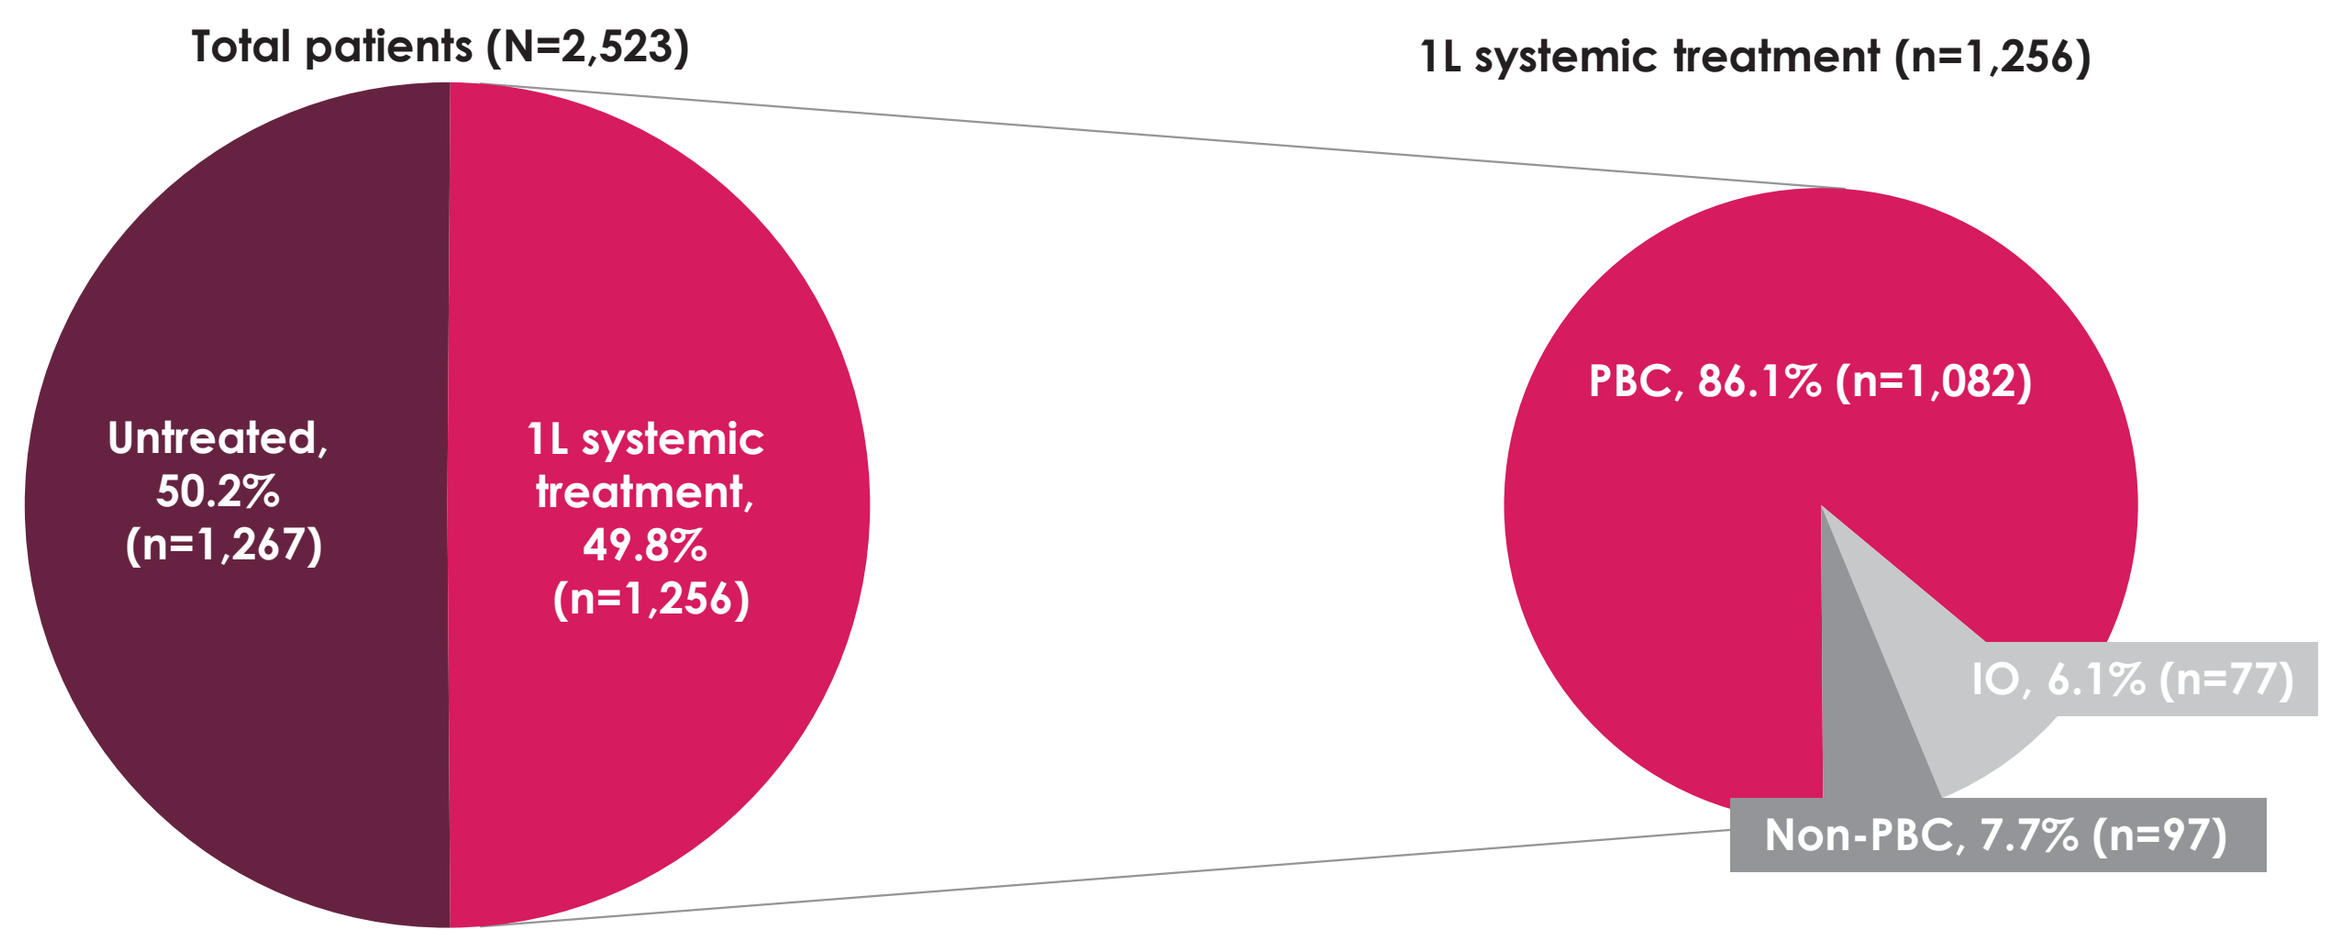
Total patients (N=2,523)
1L systemic treatment (n=1,256)
Untreated, 50.2% (n=1,267)
1L systemic treatment, 49.8% (n=1,256)
PBC, 86.1% (n=1,082)
IO, 6.1% (n=77)
Non-PBC, 7.7% (n=97)

1L, first line; **PBC**, platinum-based chemotherapy; **non-PBC**, non–platinum-based chemotherapy; **IO**, immuno-oncology.

Table 2. Most common comorbidities during the study period

Comorbidity by group of WHO codes during study period, %	Total patients (n=2,523)	Untreated (n=1,267)	1L systemic treatment (n=1,256)	1L treatment cohorts			*ICD-10* codes
				PBC (n=1,082)	Non-PBC (n=97)	IO (n=77)	
Diseases of the circulatory system	93.3	93.2	93.3	93.1	93.8	96.1	I00-I09
Diseases of the digestive system	79.4	82.3	76.4	75.9	81.4	77.9	K00-K93
Diseases of the musculoskeletal system and connective tissue	77.5	77.3	77.6	76.2	84.5	88.3	M00-M99
Diseases of the respiratory system	74.4	76.8	72.1	71.3	75.3	77.9	J00-J99
Endocrine, nutritional, and metabolic diseases	68.6	70.0	67.2	65.6	73.2	81.8	E00-E90
Diseases of the blood and blood-forming organs and certain disorders involving the immune mechanism	48.8	46.6	50.9	50.1	59.8	50.6	D50-D89
Diseases of the nervous system	28.5	32.1	24.8	23.6	25.8	41.6	G00-G99
Mental and behavioral disorders	25.2	26.4	24.0	23.9	17.5	32.5	F00-F99

1L, first line; ***ICD-10***, *International Classification of Diseases, Tenth Revision*; **IO**, immuno-oncology; **non-PBC**, non–platinum-based chemotherapy; **PBC**, platinum-based chemotherapy; **Tx**, treatment; **WHO**, World Health Organization.

Treatment trends over time

- Treatment rates and distribution of 1L systemic therapies initiated by treatment class are summarized by year in **Figure 4**
 - The proportion of treated vs untreated patients grew continously during the study period; both PBC and IO cohorts increased in size whereas the non-PBC cohort did not
 - The half-year study period for 2021 may cause uncertainty in the trends

Clinical Outcomes

- The unadjusted median OS from the index date (start date of 1L systemic treatment) was 12.8 months (95% CI, 11.5-14.1 months) with 1L PBC, 7.5 months (95% CI, 5.8-10.1 months) with 1L non-PBC, and 6.3 months (95% CI, 2.9-9.0 months) with 1L IO (**Table 3**)
- The unadjusted median OS from the index date (date of mUC diagnosis) was 7.8 months (95% CI, 6.7-8.8 months) in untreated patients (**Table 3**)
- **Figures 5** and **6** show unadjusted median OS by treatment cohort and by systemic treatment type, respectively

Figure 4. Incidence and prevalence rates over the study period (2016-2021) for the 1L-treated and untreated cohorts

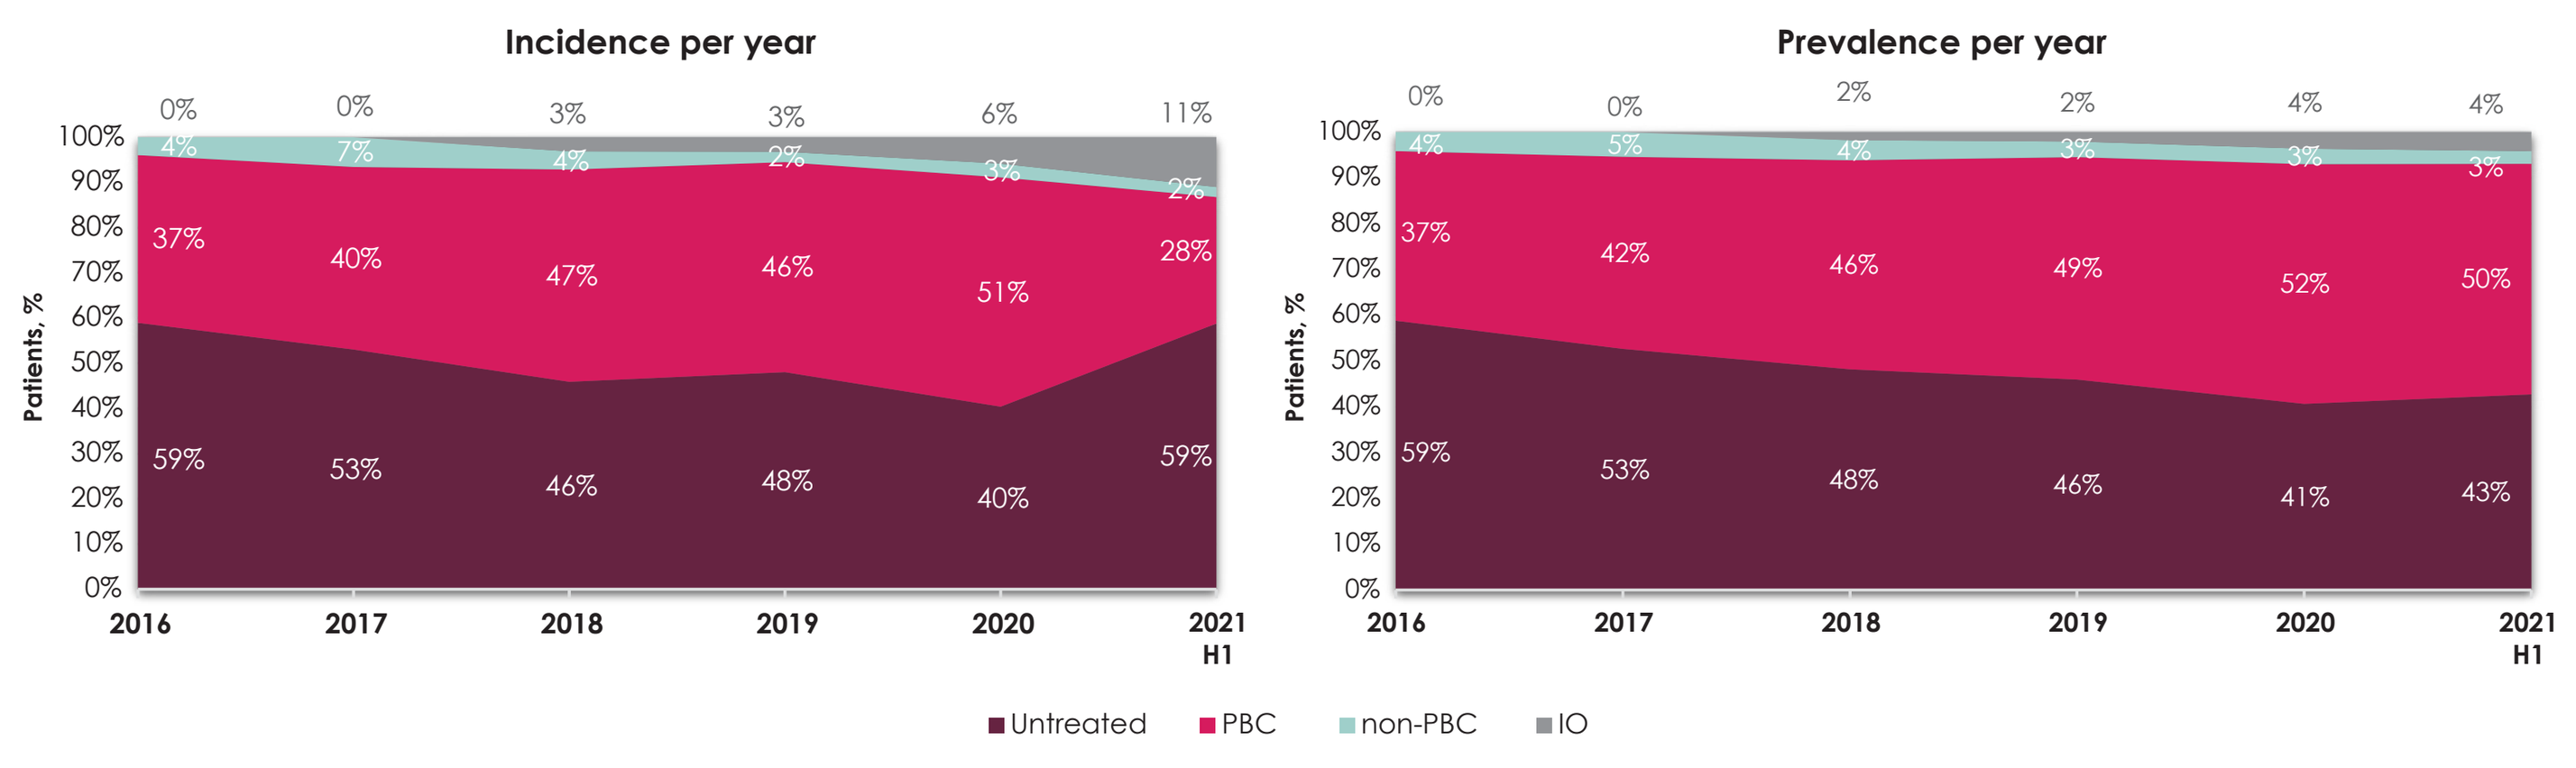
Incidence per year
Prevalence per year
Patients, %
0% 0% 3% 3% 6% 11%
4% 7% 4% 2% 3% 2%
37% 40% 47% 46% 51% 28%
59% 53% 46% 48% 40% 59%
0% 0% 2% 2% 4% 4%
4% 5% 4% 3% 3% 3%
37% 42% 46% 49% 52% 50%
59% 53% 48% 46% 41% 43%
2016 2017 2018 2019 2020 2021 H1
Untreated PBC non-PBC IO

IO use was limited as it only became available by NPBR program later in the study period (since 2018).
IO, immuno-oncology; **non-PBC**, non–platinum-based chemotherapy; **NPBR**, Named-Patient Based Reimburstment; **PBC**, platinum-based chemotherapy.

Table 3. Unadjusted median OS by cohort

Cohort	Unadjusted median OS (95% CI), months
All patients (N=2,523)	9.9 (9.2-10.7)
1L systemic treatment (n=1,256)	11.7 (10.5-12.9)
PBC (n=1,082)	12.8 (11.5-14.1)
Non-PBC chemotherapy (n=97)	7.5 (5.8-10.1)
IO (n=77)*	6.3 (2.9-9.0)
No 1L therapy (n=1,267)†	7.8 (6.7-8.8)

1L, first line; **IO**, immuno-oncology; **mUC**, metastatic urothelial carcinoma; **non-PBC**, non–platinum-based chemotherapy; **PBC**, platinum-based chemotherapy.
*Avelumab was approved by the European Commission as 1L maintenance in January 2021 and thus is not included in the study/analysis. †Index date was the date of mUC diagnosis.

Figure 5. Unadjusted median OS by patient cohort

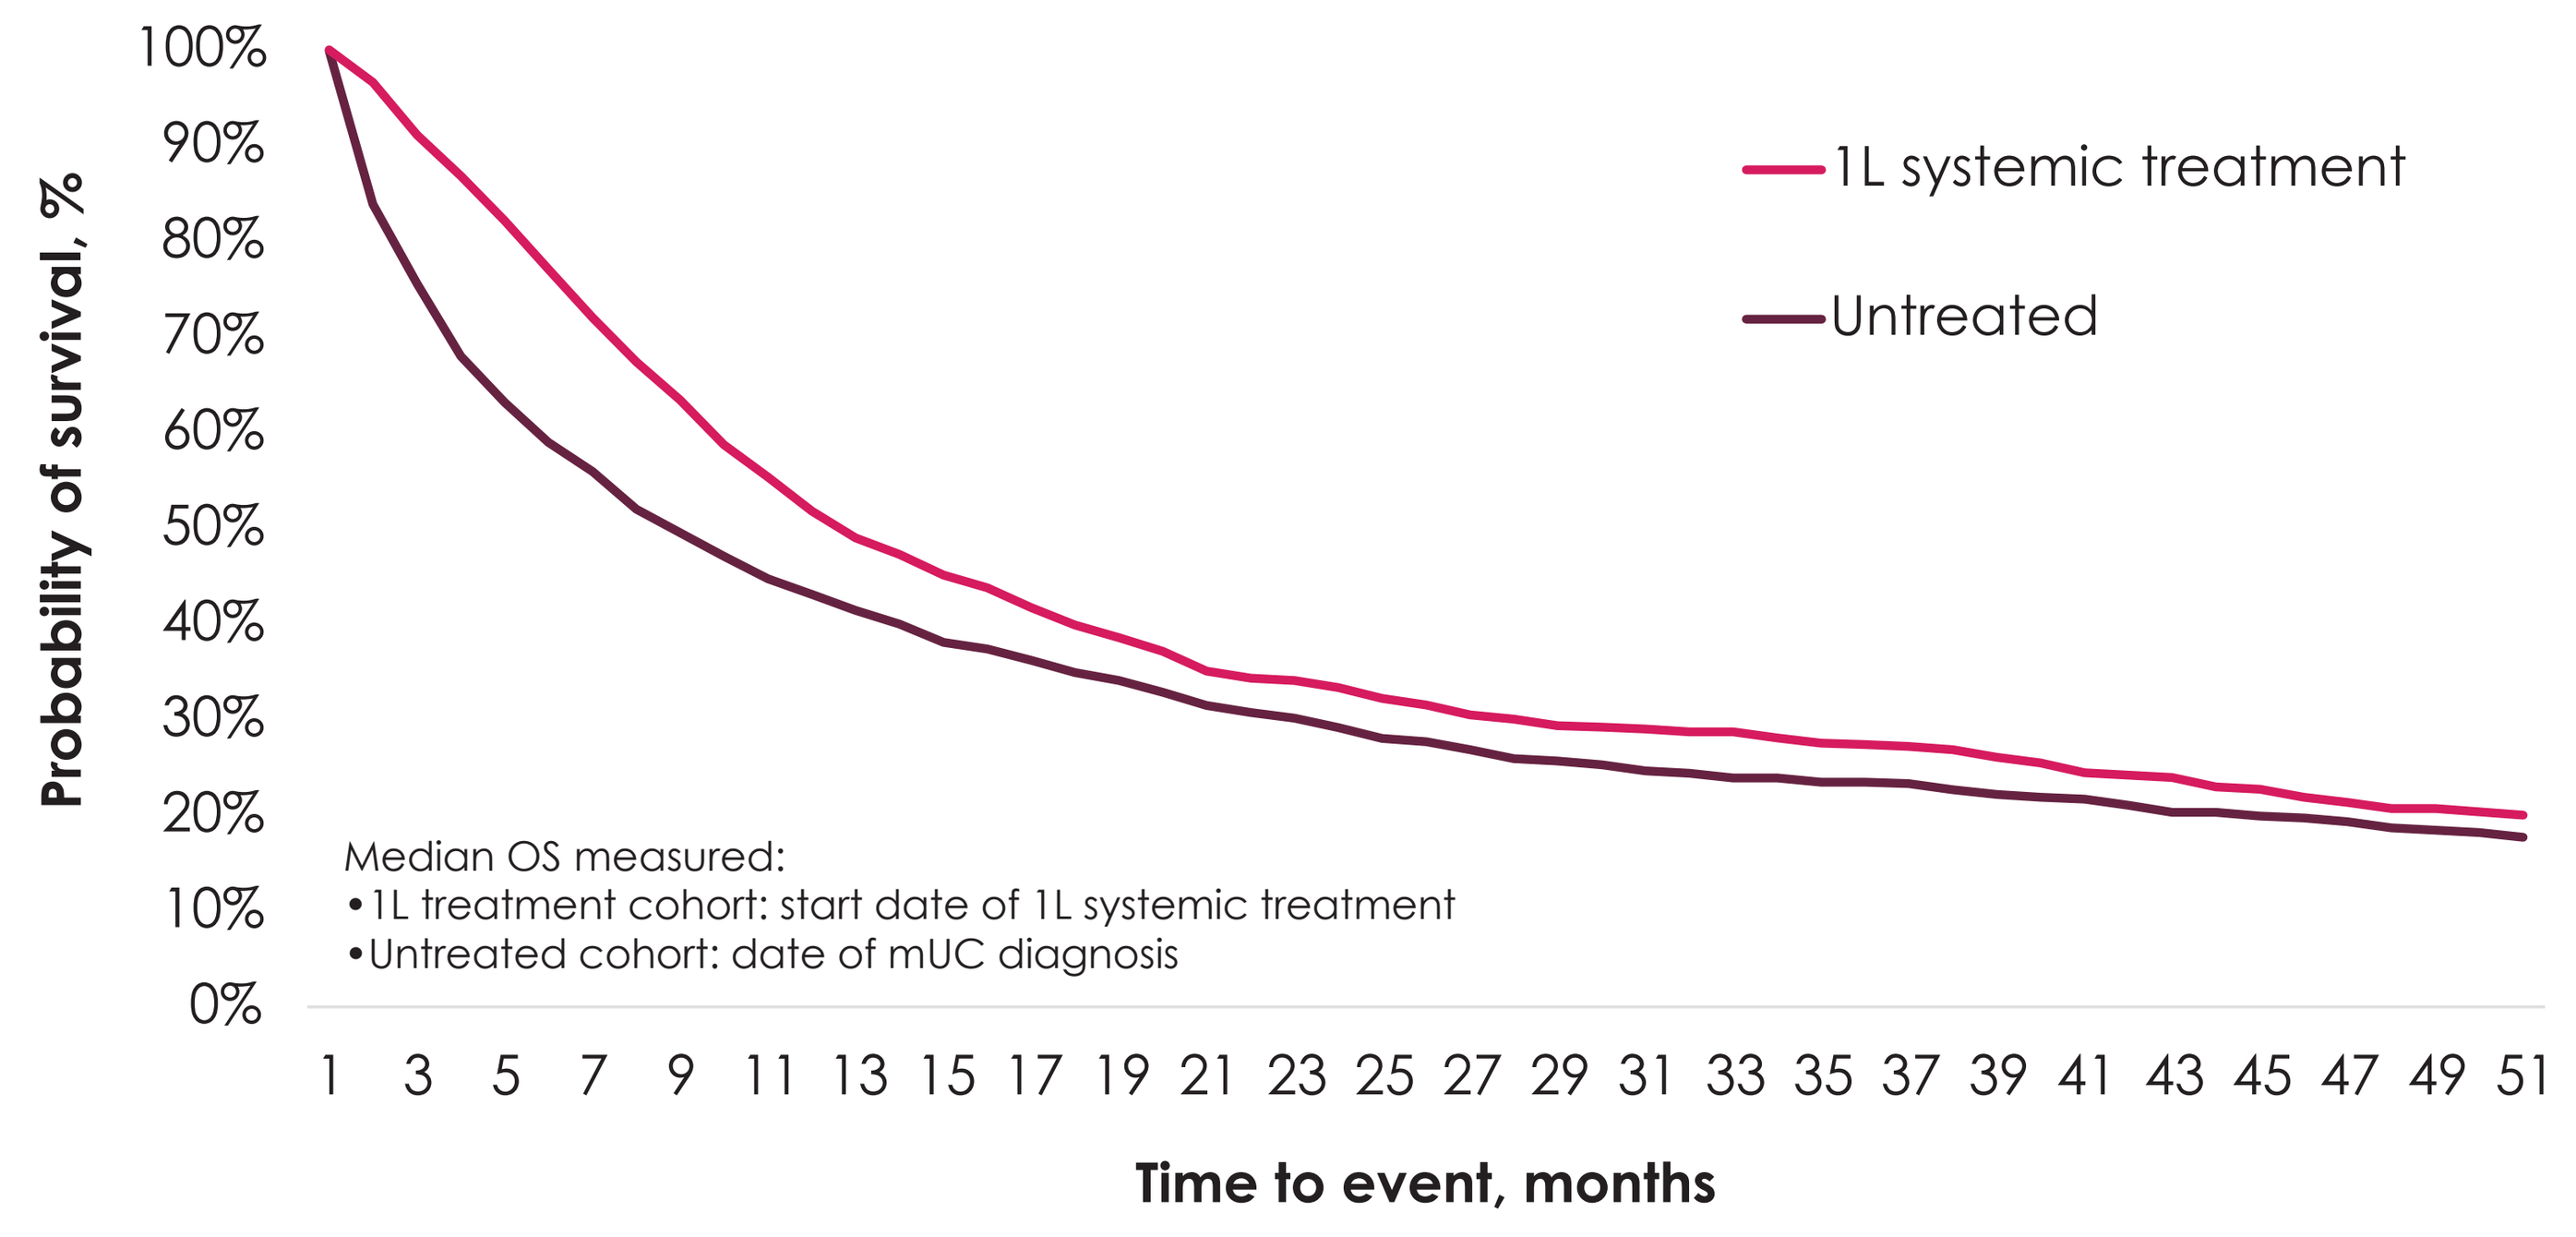
Probability of survival, %
100% 90% 80% 70% 60% 50% 40% 30% 20% 10% 0%
1L systemic treatment
Untreated
Median OS measured:
•1L treatment cohort: start date of 1L systemic treatment
•Untreated cohort: date of mUC diagnosis
1 3 5 7 9 11 13 15 17 19 21 23 25 27 29 31 33 35 37 39 41 43 45 47 49 51
Time to event, months

1L, first line; **IO**, immuno-oncology; **non-PBC**, non–platinum-based chemotherapy; **OS**, overall survival; **PBC**, platinum-based chemotherapy.

Figure 6. Unadjusted median OS by type of 1L systemic treatment

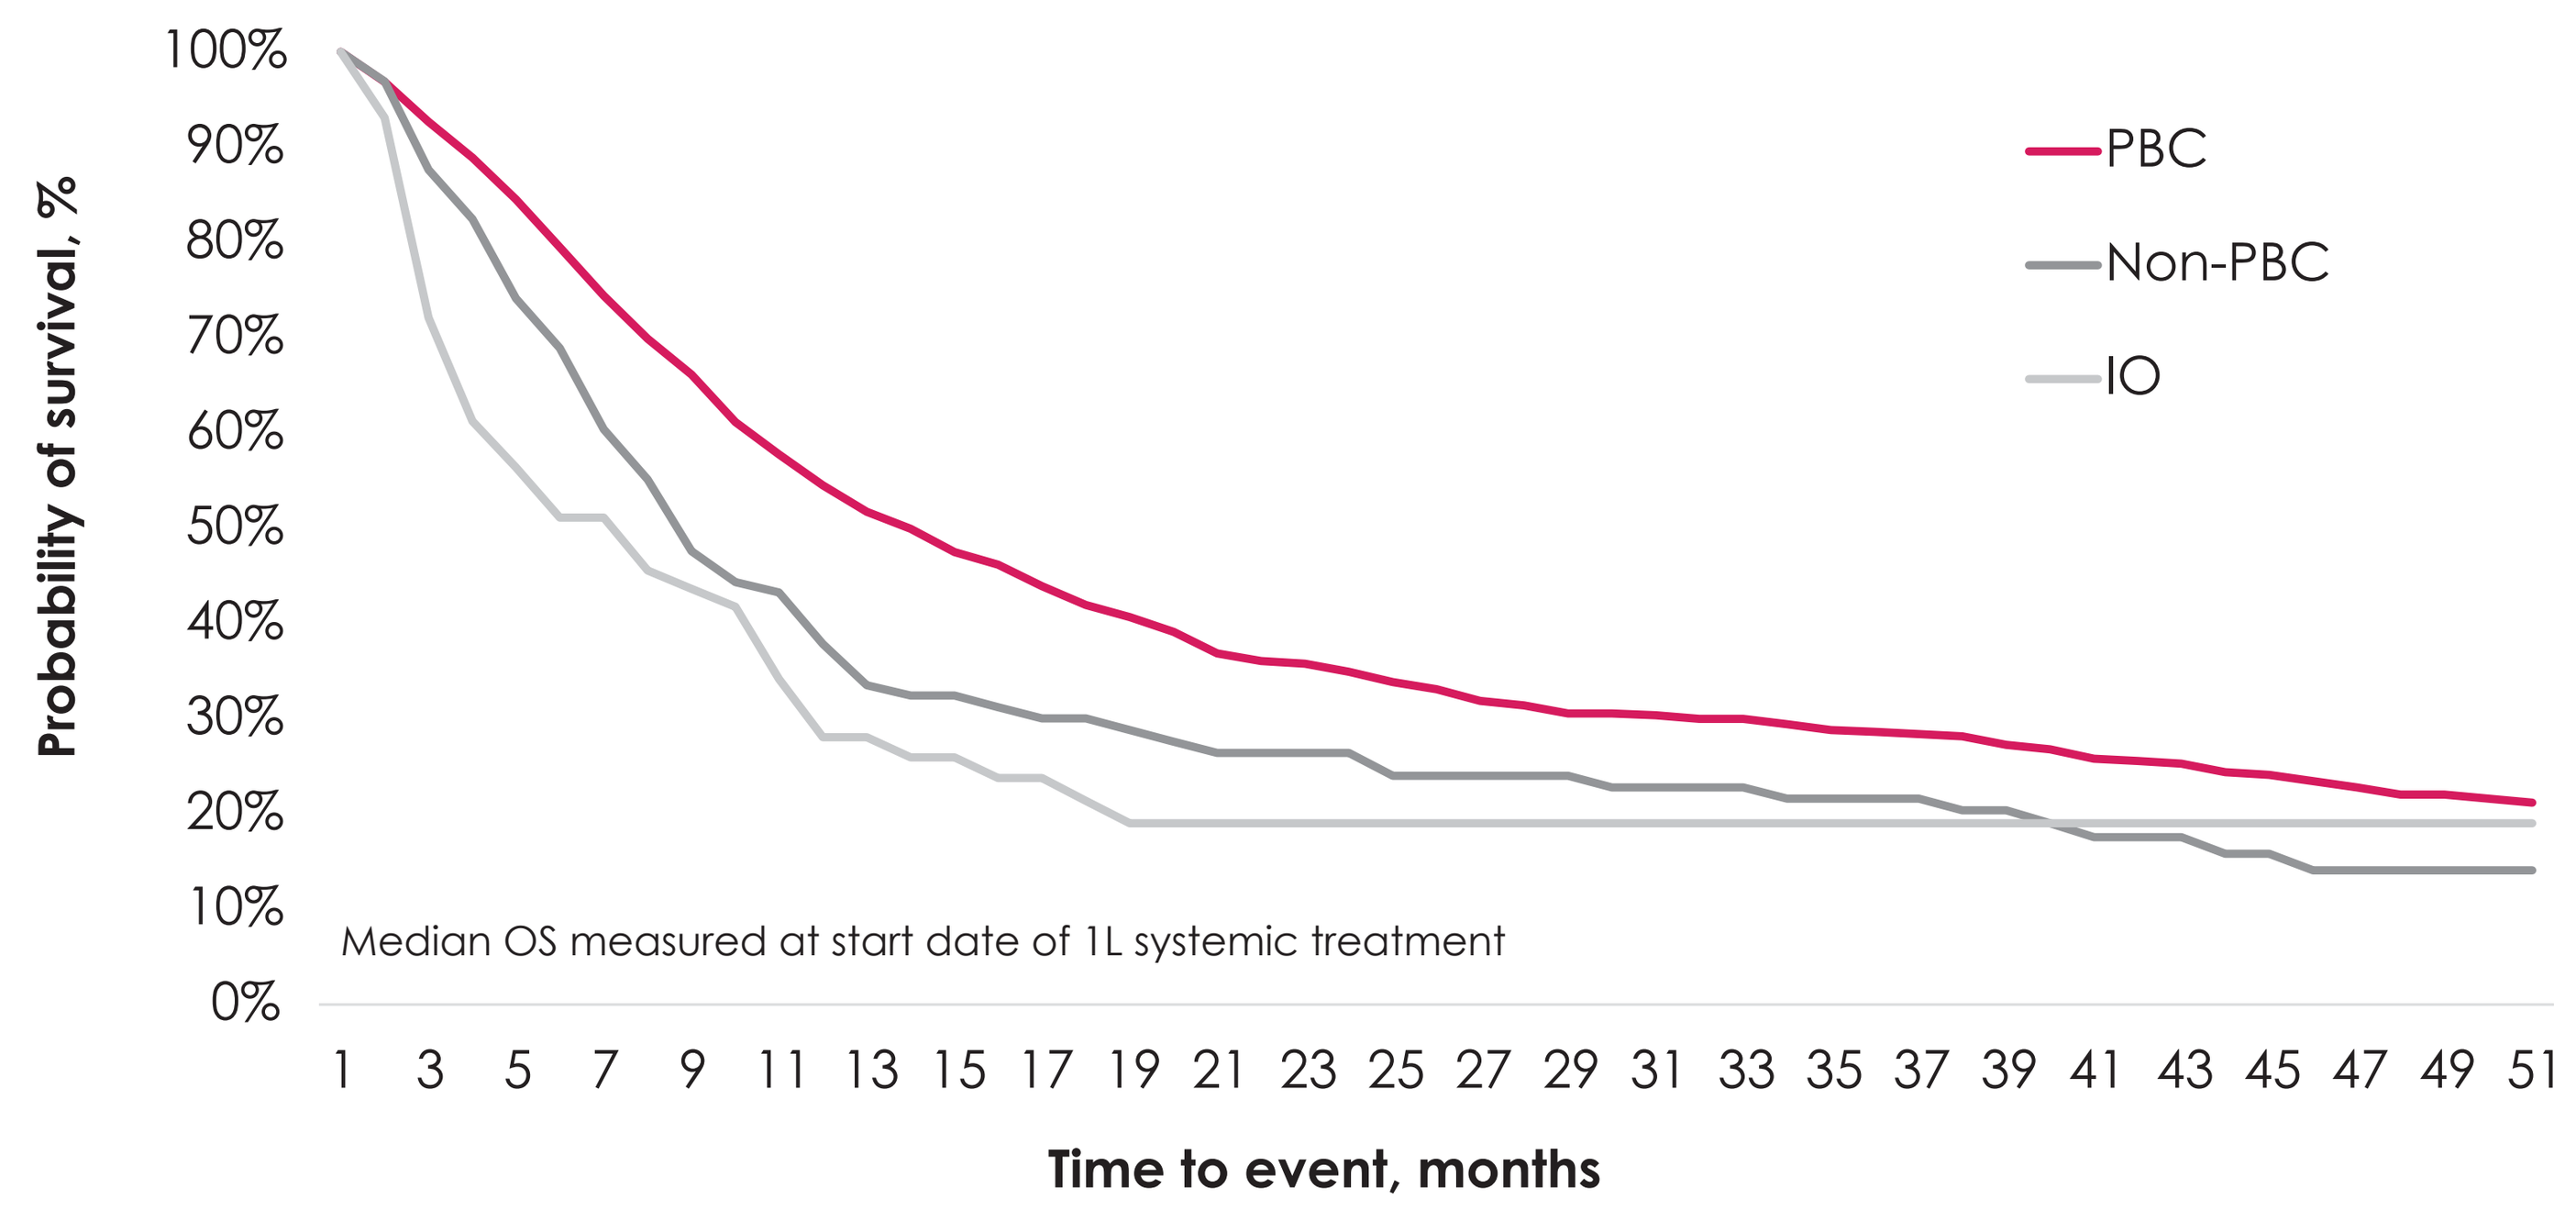
Probability of survival, %
100% 90% 80% 70% 60% 50% 40% 30% 20% 10% 0%
PBC
Non-PBC
IO
Median OS measured at start date of 1L systemic treatment
1 3 5 7 9 11 13 15 17 19 21 23 25 27 29 31 33 35 37 39 41 43 45 47 49 51
Time to event, months

1L, first line; **IO**, immuno-oncology; **non-PBC**, non–platinum-based chemotherapy; **OS**, overall survival; **PBC**, platinum-based chemotherapy.

Study limitations

- The NHIFA database lacks information on mUC disease-specific parameters, such as stage at diagnosis, treatment outcome, or known mUC risk factors, such as smoking history
- Algorithms used to identify lines of therapy based on administrative claims may not reflect the definitions of those used in clinical practice
- Due to the specificity of the secondary claims database, mUC diagnosis was identified either by *ICD-10* code or by 1L treatment
- Subgroups of <10 patients were not included in the analysis, in line with the current data protection regulations of the NHIFA
- No information was available in the claims data on the potential reasons for nonreceipt of 1L systemic treatment nor on the rationale for treatment initiation or discontinuation/switch
- The study is limited in its ability to comprehensively capture the utilization and associated outcomes of 1L IO agents

REFERENCES 1. Bilen MA, et al. Ann Oncol. 2021;32(suppl 5):S678-S724. **2.** Geynisman DM, et al. Urol Oncol. 2022;40(5):195.e1-195.e11. **3.** Dinan MA, et al. J Geriatr Oncol. 2021;12:298-304. **4.** Simeone JC, et al. Cancer Epidemiol. 2019;60:121-127. **5.** Siegel RL, et al. CA Cancer J Clin. 2019;69(1):7-34. **6.** International Agency for Research on Cancer. Accessed July 22, 2022. https://gco.iarc.fr/ **7.** Saginala K, et al. Med Sci (Basel). 2020;13;8(1):15. **8.** Bray F, et al. CA Cancer J Clin. 2018;68(6):394-424. **9.** Maráz A, et al. Magy Onkol. 2021;65:329-337. **10.** Géczi L, et al. Magy Onkol. 2021;7;65(4):339-346. **11.** Powles T, et al. Ann Oncol. 2022;33(3):244-258. **12.** Witjes JA, et al. Accessed July 14, 2022. https://uroweb.org/guideline/bladder-cancer-muscle-invasive-and-metastatic/. **13.** National Health Insurance Fund of Hungary (NEAK). Accessed July 22, 2022. http://www.neak.gov.hu/felso_menu/rolunk/kozerdeku_adatok/tevekenysegre_mukodesre_vonatkozo_adatok/a_szerv_feladata_alaptevekenysege_es_hatarkore/en_a_szerv_alaptevekenyege_feladata_es_hatarkore. **14.** ETT-TUKEB. Scientific and Research Ethics Committee. Accessed July 22, 2022. https://ett.aeek.hu/tukeb/. **DISCLOSURES A. Maráz** has served in advisory roles for MSD and Merck; and has served on speakers bureaus for MSD, Roche, Merck, and Bristol Myers Squibb. **B. Nagy, T. Macher, and J. Jeskó** received consulting fees from Merck for their work on this study. **E. Tischler and Cs. Csongvai** report employment by Merck Kft, Budapest, Hungary, an affiliate of Merck KGaA. **M. Kearney** reports employment by Merck Healthcare KGaA, Darmstadt, Germany; and reports stock and other ownership interests in Merck, Novartis, and UCB Biopharma SPRL. **ACKNOWLEDGMENTS** The authors thank the patients and their families, investigators, co-investigators, and the study teams at each of the participating centers. This research was sponsored by Merck (CrossRef Funder ID: 10.13039/100009945) as part of an alliance between Merck and Pfizer. Editorial support was provided by Katherine Quiroz-Figueroa of ClinicalThinking and was funded by Merck and Pfizer.

Poster No. 1756P. Presented at ESMO Congress 2022, 9-13 September 2022; Paris, France; Hybrid